UCSF University of California, San Francisco | Bixby Center for Global Reproductive Health

ANSIRH ADVANCING NEW STANDARDS IN REPRODUCTIVE HEALTH

Interest in and experience with IUD self-removal

Diana Foster, PhD[1] ■ Dan Grossman, MD[2] ■ David Turok, MD, MPH[3] ■ Jeffery Peipert, MD, MPH, MHA[4] ■ Linda Prine, MD[5] ■ Courtney Schreiber, MD, MPH[6] ■ Andrea Jackson, MD[1] ■ Rana Barar, MPH[1] ■ Eleanor Schwartz, MD, MS[7]

Background

Prior studies have shown that many women prefer methods of contraception that they can stop using without a clinician's assistance.[1] Previous work has shown that the idea of IUD self-removal appeals to many women.[2] We assessed the willingness and ability of women seeking IUD removal to remove it on their own and the effect of the self-removal feature on attitudes toward the IUD.

Methods

Between November 2012 and April 2013, women age 18 and older who presented to healthcare facilities in St. Louis, New York, San Francisco, Philadelphia, and Salt Lake City seeking IUD removal were asked to participate in an anonymous survey. Women were told that participation involved completing paper surveys (in English or Spanish), before and after removal, and that they would receive $30 for their participation.

The first survey collected basic socio-demographic data, data on women's reproductive history, satisfaction with their current IUD, reason for desiring IUD removal, willingness to recommend the IUD to a friend, and information hypothesized to affect successful IUD self-removal. Women were then given the option of attempting self-removal. If the woman agreed to try self-removal, the clinician left her alone to attempt the removal. If the woman declined or was unsuccessful in her attempt, the clinician performed the removal. Afterward, all participants completed a survey about their experience.

Results

326 women participated in the study *(see table)*. The majority (59%, 95%CI: 45-65%) were willing to attempt IUD self-removal. Their most common reasons were to "see if I can do it" (64%), followed by "I liked the idea of removing the IUD myself" (58%) and "I wanted to take part in research" (49%). The most common reasons for *not* attempting IUD self-removal were "I prefer to let the doctor/nurse do it" (66%), "afraid of pain" (56%), "afraid of injury" (55%) or "felt uncomfortable with the idea" (39%).

Among those who attempted self-removal, nearly one in five was successful (19%, 95%CI: 14-25%). The mean string length among this group was 6.7cm, compared to 5.4cm among those who tried and were not successful ($p<0.05$).

Over half of all women surveyed (54%) said that they were more likely to recommend the IUD to a friend, knowing that some women can remove their own IUDs; 40% said they were neither more nor less likely; and 6% said they were less likely. In a multivariate model of attitudes toward the IUD and the feature of self-removability, African American women, women who attempted self-removal, and copper IUD users were significantly more likely to recommend the IUD knowing that some women can remove their own.

Removing your own IUD

The IUD (Intrauterine Device) is in your uterus. The IUD is shaped like a T with a string attached to the bottom of the T. It has flexible arms to make inserting and removing it easier. Removal by a clinician involves pulling on the string to guide the IUD out of the uterus and vagina. We are studying whether women can remove the device themselves and how this option might affect their attitudes toward the method.

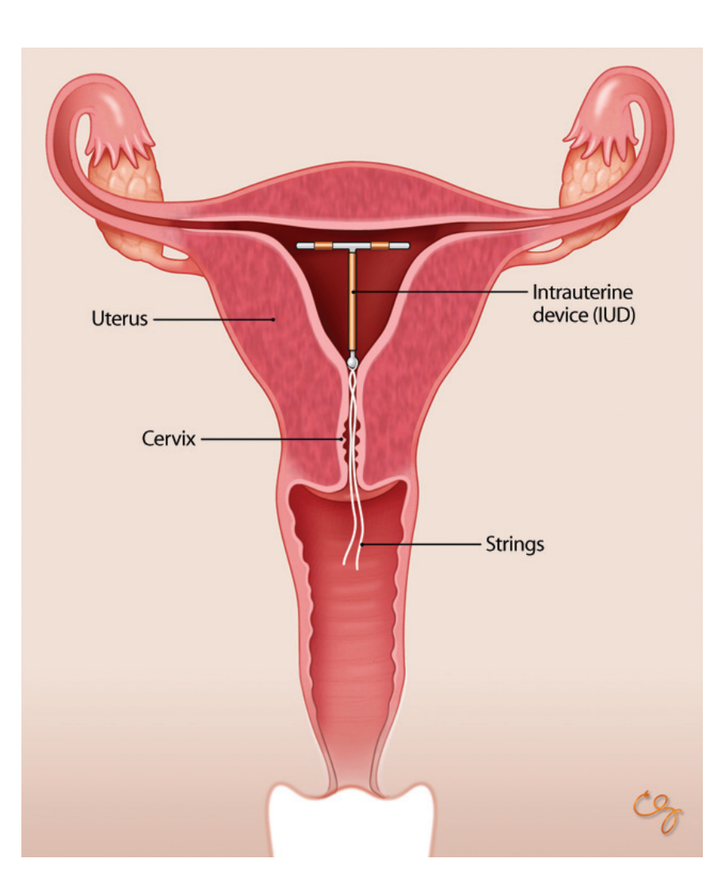
Uterus
Intrauterine device (IUD)
Cervix
Strings

Steps to remove your own IUD:

1. Wash your hands with soap.
2. Try to find a position where you can best reach the string. Here are a few suggestions:

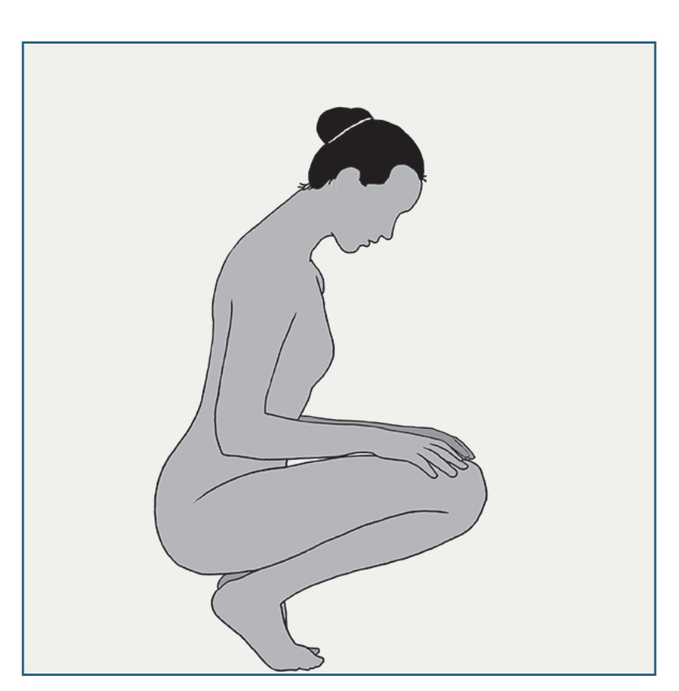

Squatting

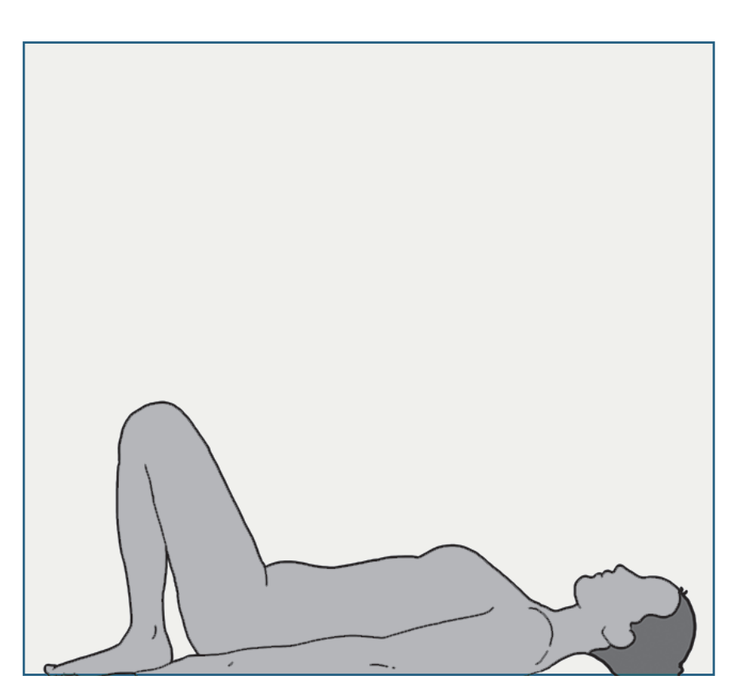

Lying down on your back with your knees up

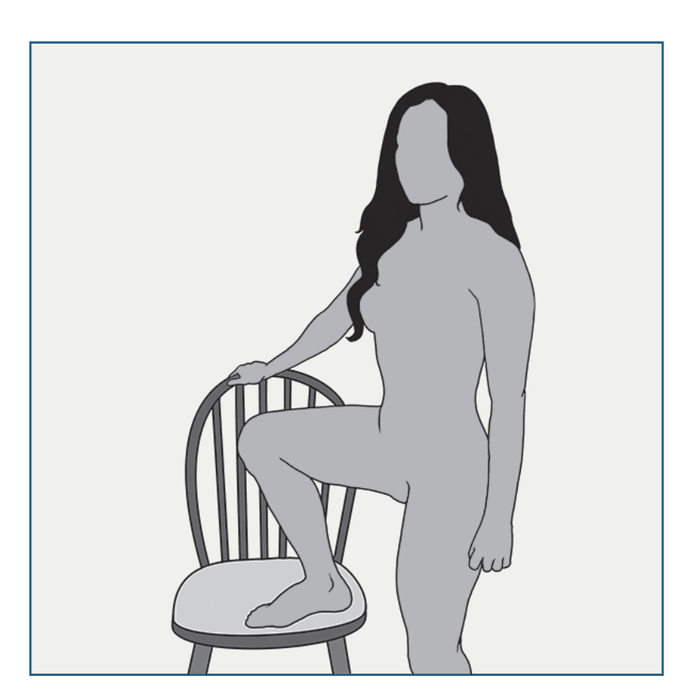

One foot up on a step or chair

3. Use your fingers to try to feel the IUD string inside your vagina. If you don't feel it, sweep your finger around your cervix and the sides of your vagina.
 - If you can touch the string, try to grasp it between two fingers.
4. Pull on the string gently but firmly. The device should come out with a gentle tug.
 - Continue pulling the string until the device comes out of your vagina.

If you aren't able to find the string, if it slips through your fingers, or if you are feeling frustrated, you can stop trying at any time. The clinician can remove it for you. **Please do not throw away your IUD. Leave it for the doctor or nurse.**

Discussion

We found that the majority of women were interested in attempting IUD self-removal, and that many reported that the self-removal option made them more likely to recommend the method to a friend. In particular, African-American women were four times more likely to recommend the IUD when the option of self-removal was presented. Given the disgraceful history of coercive sterilization in the US,[3,4] ongoing fears of reproductive coercion[5] among minority women are understandable. However, low levels of IUD use among African American women[6] likely contribute to their disproportionate burden of unintended pregnancy. Given many women's tenuous access to healthcare across the world, an effective contraceptive method where discontinuation is not dependent on a skilled clinician can help women control their fertility.

Willingness to try and success in IUD self-removal; effect on recommendation
(mixed effects logistic regression model)

	Willingness to try IUD self-removal			Success among those who try			Likelihood of recommending the IUD		
	OR	p value	95% CI	OR	p value	95% CI	OR	p value	95% CI
Age (years):									
■ 18-24 *(30% of sample)*	0.84	0.751	[0.29, 2.46]	1.56	0.691	[0.18, 13.75]	0.81	0.682	[0.29, 2.23]
■ 25-34 *(54%)*	0.53	0.145	[0.23, 1.24]	3.45	0.219	[0.48, 24.83]	1.60	0.261	[0.70, 3.65]
■ 35-49 *(16%)*									
Number of previous births	0.89	0.414	[0.68, 1.17]	0.77	0.312	[1.00, 1.00]	0.99	0.963	[0.77, 1.28]
Race/ethnicity									
■ White, non Hispanic *(41%)*									
■ Hispanic *(27%)*	1.35	0.465	[0.60, 3.01]	0.61	0.584	[0.10, 3.64]	1.41	0.360	[0.67, 2.97]
■ African American *(20%)*	1.34	0.561	[0.50, 3.54]	0.97	0.965	[0.21, 4.38]	4.38	0.000	[1.98, 9.69]
■ Asian/Other *(12%)*	2.00	0.155	[0.77, 5.22]	0.88	0.890	[0.16, 4.99]	1.36	0.479	[0.58, 3.22]
Education									
■ Some high school	1.68	0.368	[0.54, 5.19]	0.55	0.613	[0.05, 5.67]	1.89	0.244	[0.65, 5.49]
■ High school diploma/GED	1.31	0.554	[0.54, 3.21]	0.90	0.914	[0.14, 5.68]	0.88	0.779	[0.38, 2.08]
■ Some college/technical school	1.05	0.907	[0.47, 2.34]	1.75	0.462	[0.39, 7.75]	1.83	0.125	[0.84, 3.97]
■ Bachelor degree									
Obesity									
■ <30 BMI									
■ 30-40	0.84	0.61	[0.44, 1.62]	0.29	0.076	[0.07, 1.14]	0.87	0.678	[0.47, 1.65]
■ >40	0.80	0.697	[0.25, 2.51]	5.95	0.148	[0.53, 66.71]	0.99	0.991	[0.32, 3.08]
Nativity									
■ Born in the US									
■ Born in another country	0.40	0.023	[0.18, 0.88]	0.82	0.829	[0.13, 5.00]	1.82	0.144	[0.82, 4.05]
Pelvic discomfort									
■ No									
■ Yes	0.63	0.208	[0.30, 1.30]	3.03	0.184	[0.59, 15.57]	0.56	0.110	[0.27, 1.14]
Type of IUD									
■ Levonorgestrel IUD									
■ Copper T IUD	1.63	0.131	[0.86, 3.06]	1.72	0.369	[0.53, 5.64]	2.03	0.027	[1.09, 3.80]
Felt string									
■ No, never tried									
■ No, couldn't feel them	2.11	0.072	[0.93, 4.74]	1.98	0.55	[0.21, 18.81]	1.24	0.587	[0.57, 2.71]
■ Yes	2.10	0.016	[1.15, 3.83]	6.47	0.021	[1.33, 31.44]	1.48	0.206	[0.81, 2.72]
Time with IUD (months)	1.01	0.378	[0.99, 1.02]	0.99	0.262	[0.97, 1.01]	1.01	0.059	[1.00, 1.02]
Total length of the string (cm)				1.63	0.002	[1.19, 2.22]			
Position attempted									
■ one foot up on a step				0.20	0.009	[0.06, 0.68]			
■ squatting				0.52	0.263	[0.17, 1.63]			
■ lying on back with knees up				0.32	0.078	[0.09, 1.14]			
■ other				8.58	0.081	[0.77, 95.65]			
Attempt to remove own IUD									
■ did not attempt									
■ try and fail							2.90	0.000	[1.63, 5.15]
■ try and succeed							1.74	0.258	[0.67, 4.53]

Note: Numbers in orange indicate statistical significance

Affiliations: [1]UCSF [2]Ibis [3]Univ. of Utah [4]Washington Univ. [5]Inst. for Family Health [6]Univ.of Pennsylvania [7]Univ.of of Pittsburgh

References: [1]Lessard et al, 2012 [2]Foster et al, 2012 [3]Stern, 2005 [4]Kluchin, 2004 [5]Kaye et al, 2009 [6]Jones et al, 2012